AAAAI Annual Meeting, Houston , TX
February 20-24, 2015

RESULTS OF A PHASE III TRIAL IN PATIENTS WITH PIDD USING AN IVIG CONTAINING HIGH TITER NEUTRALIZING ANTIBODY TO RESPIRATORY SYNCYTIAL VIRUS (RSV)

286

J Mond[1], W Du[2], A Kobayashi[3], R Kobayashi[4], C Cunningham-Rundles[5], W Lumry[6], J Harris III[7], R Levy[8], M Stein[9], LR Forbes[10], I Melamed[11], K Kestenberg[1], and R L Wasserman[12]

[1]ADMA Biologics, [2]Clinical Statistics Consulting, [3] Midlands Pediatrics, [4]Department of Pediatrics UCLA School of Medicine/Division of Pediatric Immunology and Allergy, [5]The Immunology Institute, Icahn School of Medicine at Mount Sinai, [6]AARA Research Center, [7]The South Bend Clinic, LLP, [8]Family Allergy & Asthma Center, PC, [9]Allergy Associates of the Palm Beach, P.A. , [10]Center for Human Immunobiology, Texas Childrens Hospital, [11]IMMUNOe, [12]Dallas Allergy Immunology

Introduction

Replacement therapy with Immunoglobulin G purified from pools of plasma from multiple donors has been used as intravenous immunoglobulin (IGIV) and subcutaneous injections to prevent serious infection in patients with immune deficiencies. While Ig effectively replaces the absent humoral antibody compartment of these patients, available concentrations of specific anti-pathogen antibodies in the Ig may sometimes be limiting particularly when the offending organism is presented in overwhelming numbers. We have recently completed a phase III trial in 59 subjects with PIDD, with the primary objective of preventing acute serious bacterial infections, using the investigational product RI-002, an IVIG derived from plasma donors selected for their high titers of neutralizing antibodies to RSV.

1. Patient Demographics

Characteristic/Statistics	Total (Subjects=59)	3-Week Cycle (Subjects=19)	4-Week Cycle (Subjects=40)
Age (years)			
N	59	19	40
Mean (SE)	41.8 (2.84)	38.6 (4.94)	43.3 (3.48)
SD	21.78	21.53	22.01
Min, Max	3, 74	5, 70	3, 74
Age Group			
2-6 years	2 (3.4%)	1 (5.3%)	1 (2.5%)
7-11 years	4 (6.8%)	1 (5.3%)	3 (7.5%)
12-16 years	5 (8.5%)	1 (5.3%)	4 (10.0%)
>16 years	48 (81.4%)	16 (84.2%)	32 (80.0%)
Sex			
Male	28 (47.5%)	7 (36.8%)	21 (52.5%)
Female	31 (52.5%)	12 (63.2%)	19 (47.5%)

2. Adverse Events of Interest

AE of Interest (AEOI) *	Total Subjects (N=59) n (%)	3 Week Cycle Subjects (N=19) n (%)	4 Week Cycle Subjects (N=40) n (%)
Subjects with >=1 AEOI	46 (78.0)	14 (73.7)	32 (80.0)
Bronchitis	12 (20.3)	1 (5.3)	11 (27.5)
Chronic + acute sinusitis	24 (40.7)	9 (47.4)	15 (37.5)
Otitis media	1 (1.7)	0	1 (1.7)
Influenza	5 (8.5)	2 (10.5)	3 (7.5)
Nasopharyngitis	16 (27.1)	7 (36.8)	9 (22.5)
URI	20 (33.9)	6 (31.6)	14 (35.0)

AEOI used the following definitions:
1) Bronchitis included AE preferred term of Bronchitis.
2) Chronic + acute sinusitis included AE preferred terms of Acute sinusitis and Sinusitis.
3) Influenza included AE preferred terms of Influenza and H1N1 influenza.
4) Nasopharyngitis included AE preferred terms of Nasopharyngitis, Pharyngitis streptococcal, and Viral pharyngitis.
5) URI included AE preferred terms of Croup infectious,Upper respiratory tract infection, and Viral upper respiratory tract infection.

3. Serious Adverse Events

SAE	Total: Total Events [1]	Total: Subjects (N=59) n (%) [2]	Total: Infusions (I=793) n (%) [3]	3-Week Cycle: Total Events [1]	3-Week Cycle: Subjects (N=19) n (%) [2]	3-Week Cycle: Infusions (I=294) n (%) [3]	4-Week Cycle: Total Events [1]	4-Week Cycle: Subjects (N=40) n (%) [2]	4-Week Cycle: Infusions (I=499) n (%) [3]
Serious Bacterial Infection	0	0	0	0	0	0	0	0	0
Postoperative wound infection	1	1 (1.7)	1 (0.1)	0	0	0	1	1 (2.5)	1 (0.2)
Migraine	1	1 (1.7)	1 (0.1)	0	0	0	1	1 (2.5)	1 (0.2)

4. Summary of Unscheduled Medical Visits

Summary Category	Total (Subjects=59)	3-Week Cycle (Subjects=19)	4-Week Cycle (Subjects=40)
Unscheduled Medical Visits Due to Infection			
Total Visits in the study	54	18	36
Visits per person per year	0.966	1.041	0.933
1-Sided 95% Upper Bound	1.209	1.533	1.227
Unscheduled Medical Visits for Other Reasons			
Total Visits in the study	40	12	28
Visits per person per year	0.716	0.694	0.726
1-Sided 95% Upper Bound	0.928	1.115	0.990

5. Summary of Hospitalizations

Summary Category	Total (Subjects=59)	3-Week Cycle (Subjects=19)	4-Week Cycle (Subjects=40)
Total Hospitalizations due to infection	1	0	1
Number of days of Hospitalization	5	0	5
Hospitalizations per subject per year	0.018	0.000	0.026

6. Number of Days Lost from Work/School/Day Care Due to Infections and their Treatment

Statistics	Total (Subjects=59)	3-Week Cycle (Subjects=19)	4-Week Cycle (Subjects= 40)
Total days missed in the study	93	27	66
No. of days missed per person per year	1.66	1.56	1.71
1-sided 95% Upper Bound	1.97	2.14	2.09

7. Summary of Study Drug Related TEAEs[1] Occuring in ≥ 5% of Subjects

Preferred Term	Total: Total Events	Total: Subjects (N=59) n (%)	Total: Infusions (I=793) n (%)	3-Week Cycle: Total Events	3-Week Cycle: Subjects (N=19) n (%)	3-Week Cycle: Infusions (I=294) n (%)	4-Week Cycle: Total Events	4-Week Cycle: Subjects (N=40) n (%)	4-Week Cycle: Infusions (I=499) n (%)
Subjects / Infusions with ≥ 1 Event	55	26 (44.1)	39 (4.9)	22	10 (52.6)	17 (5.8)	33	16 (40.0)	22 (4.4)
Headache	10	8 (13.6)	10 (1.3)	3	3 (15.8)	3 (1.0)	7	5 (12.5)	7 (1.4)
Adverse drug reaction	4	3 (5.1)	4 (0.5)	0	0	0	4	3 (7.5)	4 (0.8)
Diarrhoea	3	3 (5.1)	3 (0.4)	2	2 (10.5)	2 (0.7)	1	1 (2.5)	1 (0.2)
Fatigue	7	3 (5.1)	7 (0.9)	6	2 (10.5)	6 (2.0)	1	1 (2.5)	1 (0.2)
Migraine	3	3 (5.1)	3 (0.4)	1	1 (5.3)	1 (0.3)	2	2 (5.0)	2 (0.4)
Myalgia	4	3 (5.1)	4 (0.5)	3	2 (10.5)	3 (1.0)	1	1 (2.5)	1 (0.2)
Pruritus	3	3 (5.1)	3 (0.4)	0	0	0	3	3 (7.5)	3 (0.6)

1.) TEAE- Treatment Emergent Adverse Event

8. Summary of TAAEs[1]

	Total (Subjects=59)	3-Week Cycle (Subjects=19)	4-Week Cycle (Subjects=40)
Number of Infusions with TAAEs	113	36	77
Proportion of Infusions with TAAEs	0.142	0.122	0.154
Subjects with No TAAE	16 (27.1%)	4 (21.1%)	12 (30.0%)
Subjects with <10 Infusions with TAAEs	43 (72.9%)	15 (78.9%)	28 (70.0%)

1.) TAAE- Temporally Associated Adverse Events: Adverse events that occur within 72 hours of product infusion

9. Days on Therapeutic Antibiotics

	Total (Subjects=59)	3-Week Cycle (Subjects=19)	4-Week Cycle (Subjects= 40)
Total Days on Antibiotic Therapy (A)	1839	713	1126
Total person-years (B)	55.88	17.30	38.58
Days on Antibiotic Therapy per person per year (A/B)	32.912	41.219	29.187

10. Maximum Fold Increase in Serum Antibody Neutralizing Titers to RSV, H. Influenza and S. Pneumoniae Achieved in Populations Receiving RI-002 as Compared to their Baseline Titer Prior to RI-002 Administration

		Total (n=31)*
RSV	Mean Fold Increase (s.e)	5.3 (0.54)
	P value	.0001
H. Influenza	Mean Fold Increase (s.e)	3.2 (0.25)
	P value	.0001
S. Pneumoniae	Mean Fold Increase (s.e)	2.89 (0.30)
	P value	.006

*Data on PK Subjects

Summary

RI-002 is a 10% Human immunoglobulin (IVIG) prepared with standardized elevated levels of naturally occurring anti-RSV neutralizing antibodies which was evaluated in a pivotal Phase III trial in patients diagnosed with Primary Immune Deficiency Disease (PIDD). In the trial, RI-002 successfully met its primary and secondary efficacy and safety objectives. The trial enrolled subjects ranging in age from 3-73 years, 59 patients were included in the mITT population, 55.9 total patient years were used for evaluation of the data and there were a total of 793 infusions of RI-002 during the trial. There were zero (0) serious bacterial infections and a total of only one (1) SAE due to infection for the duration of the trial. Other key data from the trial includes: only 1.66 days per subject per year were lost from work, school or day care due to infection. There were only 0.097 days per subject per year of unscheduled visits to the physician or ER, and only one (1) hospitalization of a five (5) day duration secondary to a local wound infection. The safety profile of RI-002 was comparable to that of other commercially available IVIG's.

Conclusion

RI-002 is an IVIG prepared to meet the FDA standards for immunoglobulins and contains standardized, elevated levels of RSV neutralizing antibodies. The trial has determined that RI-002 is an effective treatment for patients diagnosed with PIDD. RI-002 may provide a promising alternative to currently marketed immunoglobulins for certain patients with PIDD.

Future Investigations

In view of the low incidence of days lost from work, school or day care, minimal number of days of unscheduled visits to the physician or ER, only one reported case of otitis media in the treated study subjects as well as other favorable outcomes resulting from the secondary endpoint analysis, further studies may be warranted to explore whether the unique antibody profile of RI-002 may be correlated to these outcomes.

Figure 1
Mean (+/- SE) Total IgG Trough Levels by Infusion Number mITT Analysis Set

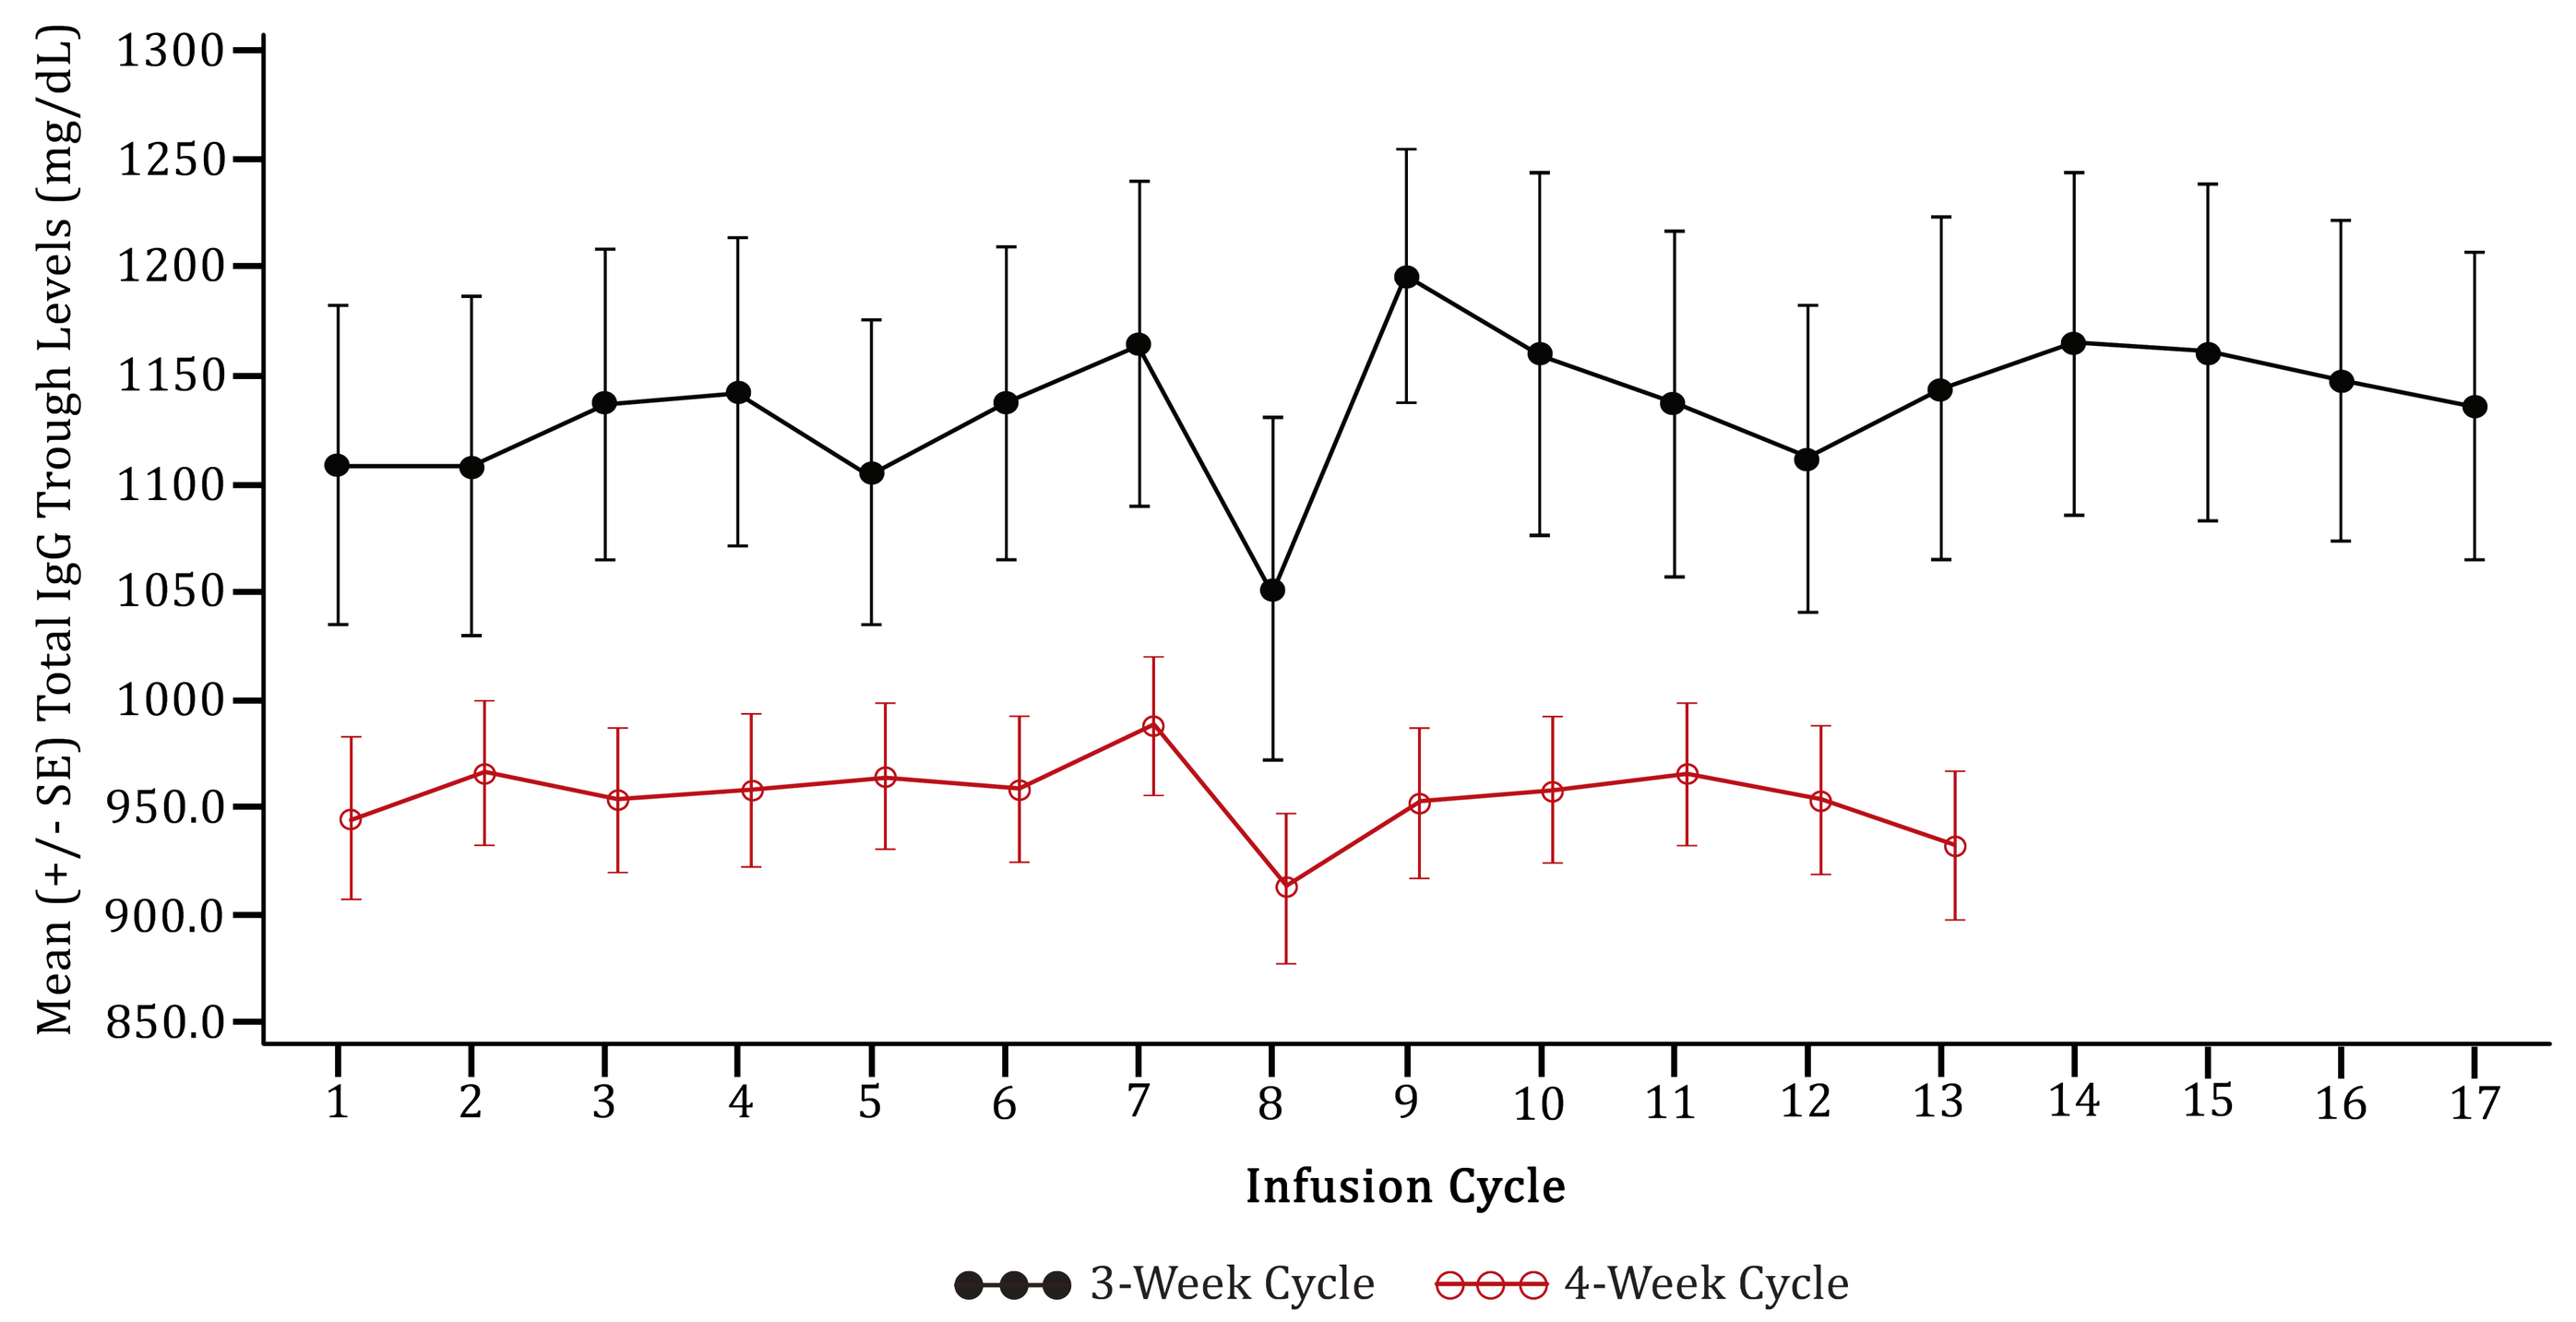
Mean (+/- SE) Total IgG Trough Levels (mg/dL)
1300
1250
1200
1150
1100
1050
1000
950.0
900.0
850.0
1 2 3 4 5 6 7 8 9 10 11 12 13 14 15 16 17
Infusion Cycle
3-Week Cycle
4-Week Cycle

Patients received on average 500 mg/kg of RI-002